Reversible immortalization of human adipose tissue-derived mesenchymal stem cells

Péter Tátrai[1,2], Áron Szepesi[3], Anna Szigeti[3], Katalin Német[1,3]

[1]Experimental Gene Therapy Lab, National Blood Transfusion Service, Budapest, Hungary; [2]Department of Biochemistry and Molecular Biology, University of Debrecen, Debrecen, Hungary; [3]Creative Cell Ltd, Budapest, Hungary

Introduction. Mesenchymal stem cells (MSCs) can be harvested from human adipose tissue and differentiated into bone, cartilage, and fat, as well as towards a putative endothelial phenotype. Our current interest is the stimulation of osteogenic and endothelial differentiation of MSCs for use in tissue regeneration research. However, heterogeneity between donors, dependence of cellular properties on passage number, and limited life span of *in vitro* MSC cultures present major hurdles for reproducible experiments.

Therefore, we aimed to establish immortalized MSC populations with well-characterized properties that can provide a steady supply of homogeneous cells for *in vitro* work.

Panels: see text for explanation

Wild type MSC

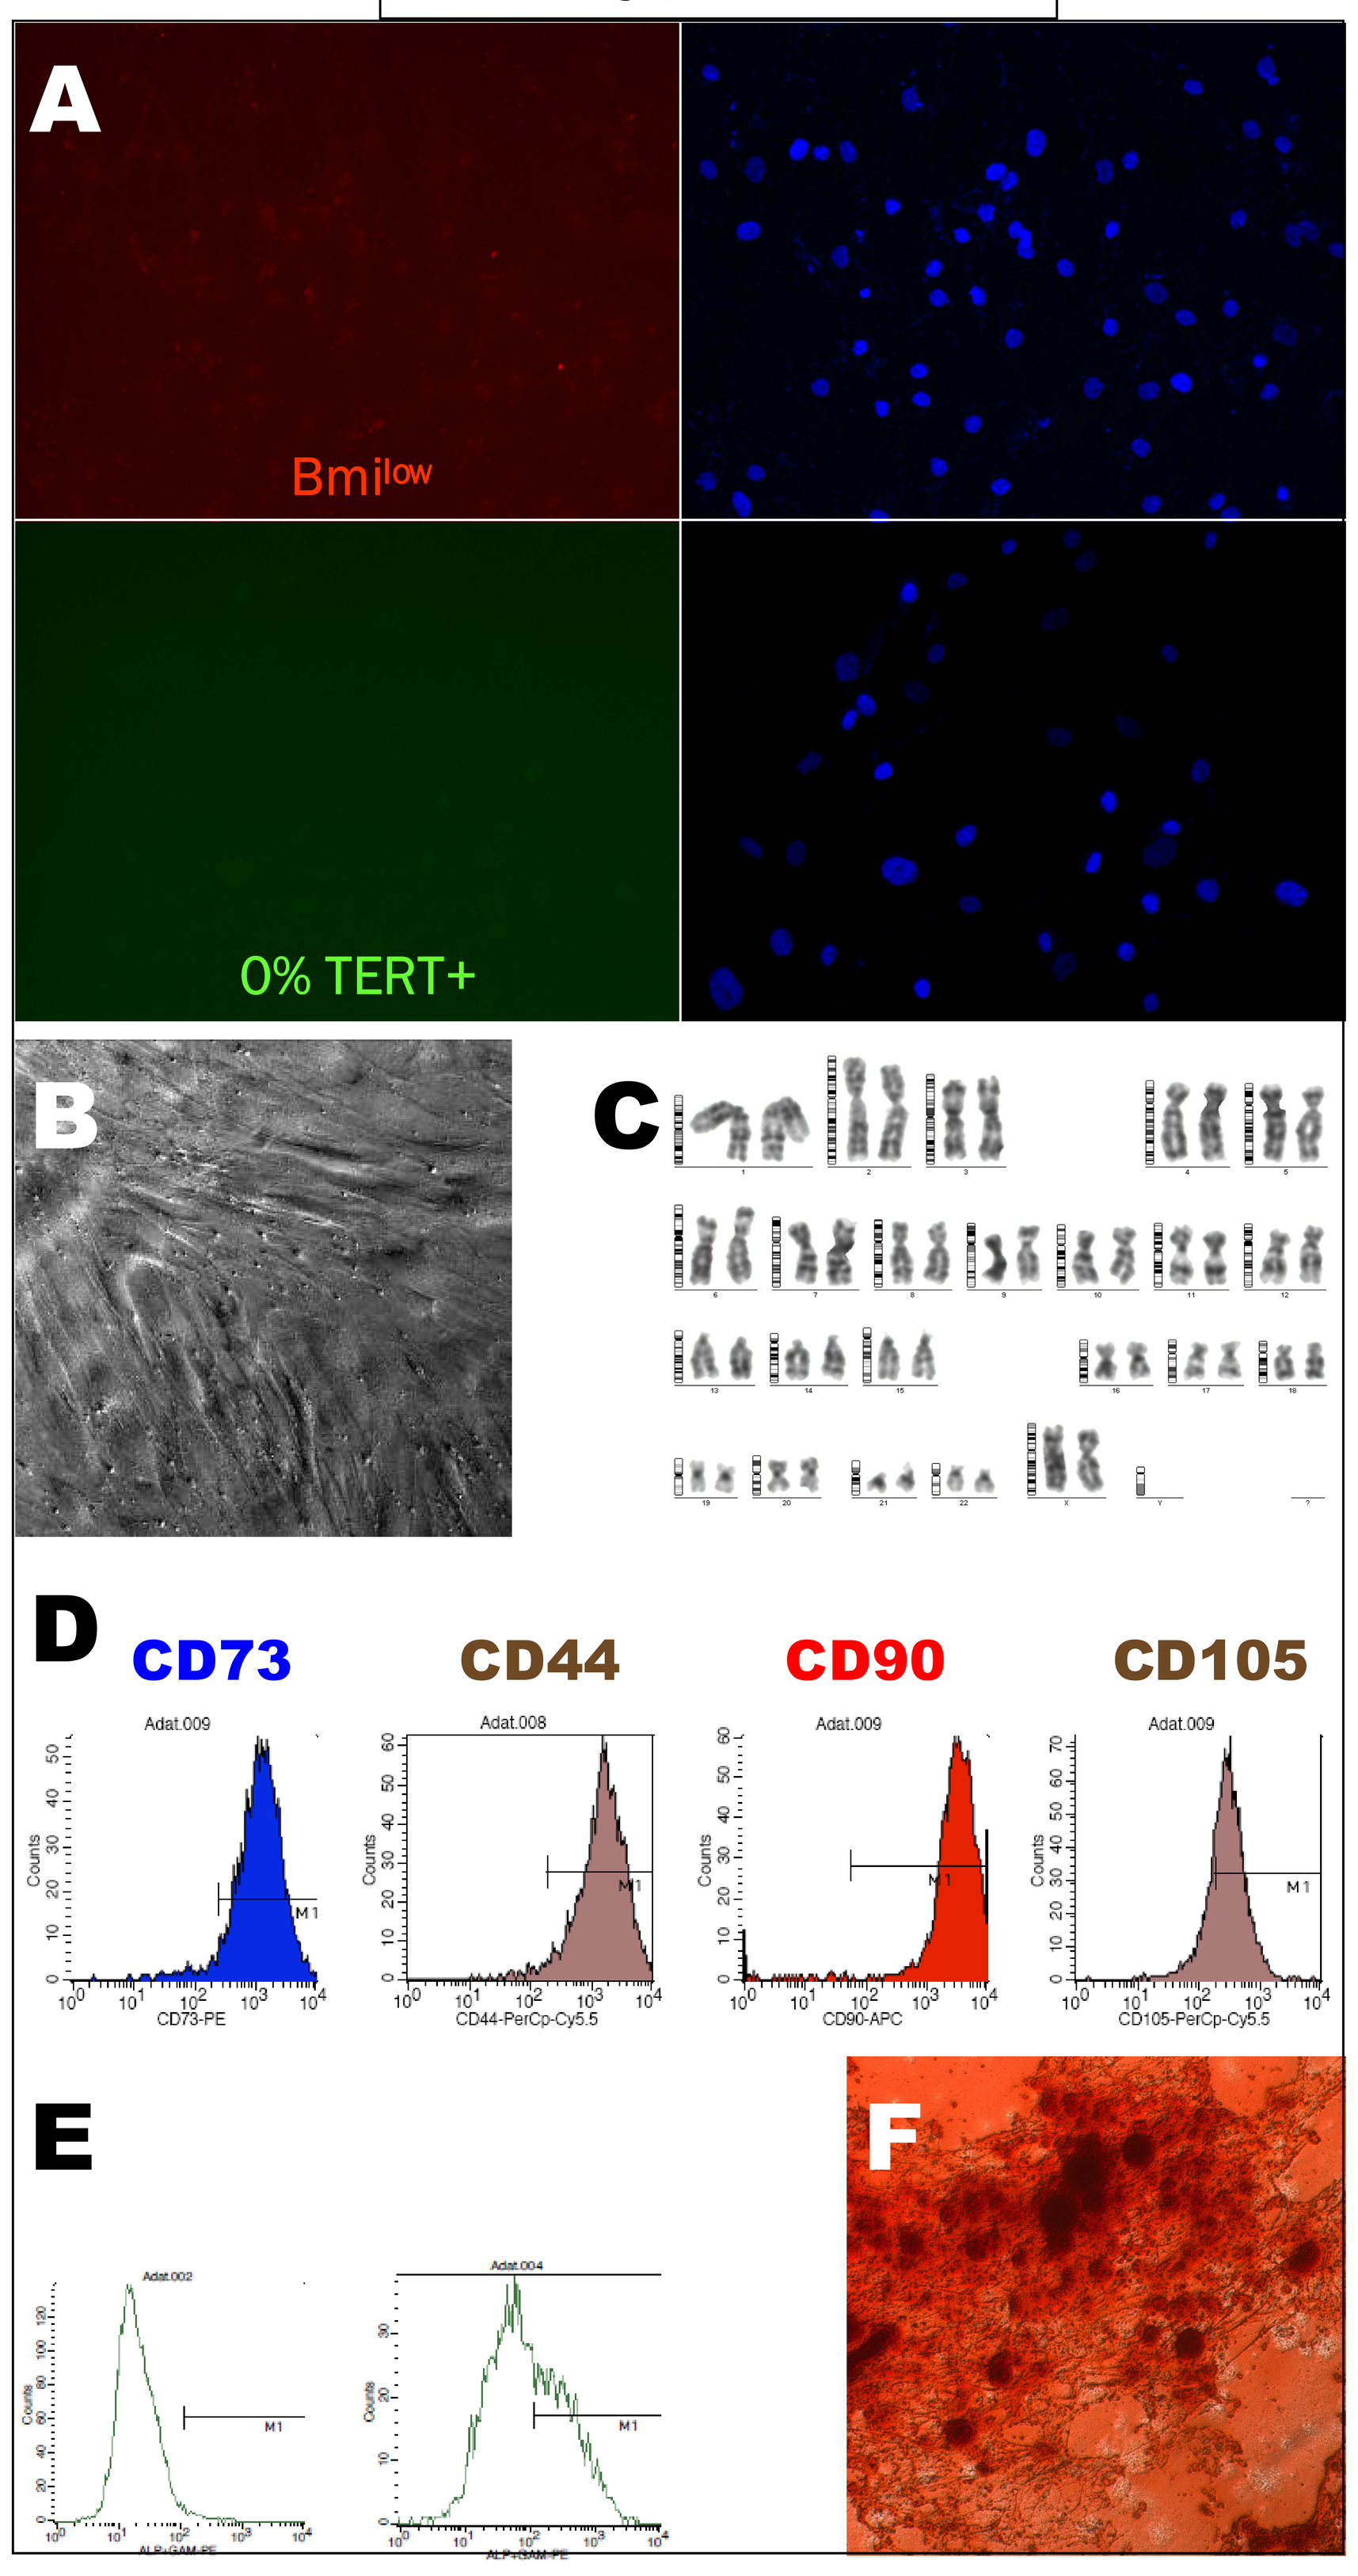
A
Bmi^low
0% TERT+
B
C
D
CD73
CD44
CD90
CD105
E
F

TERT

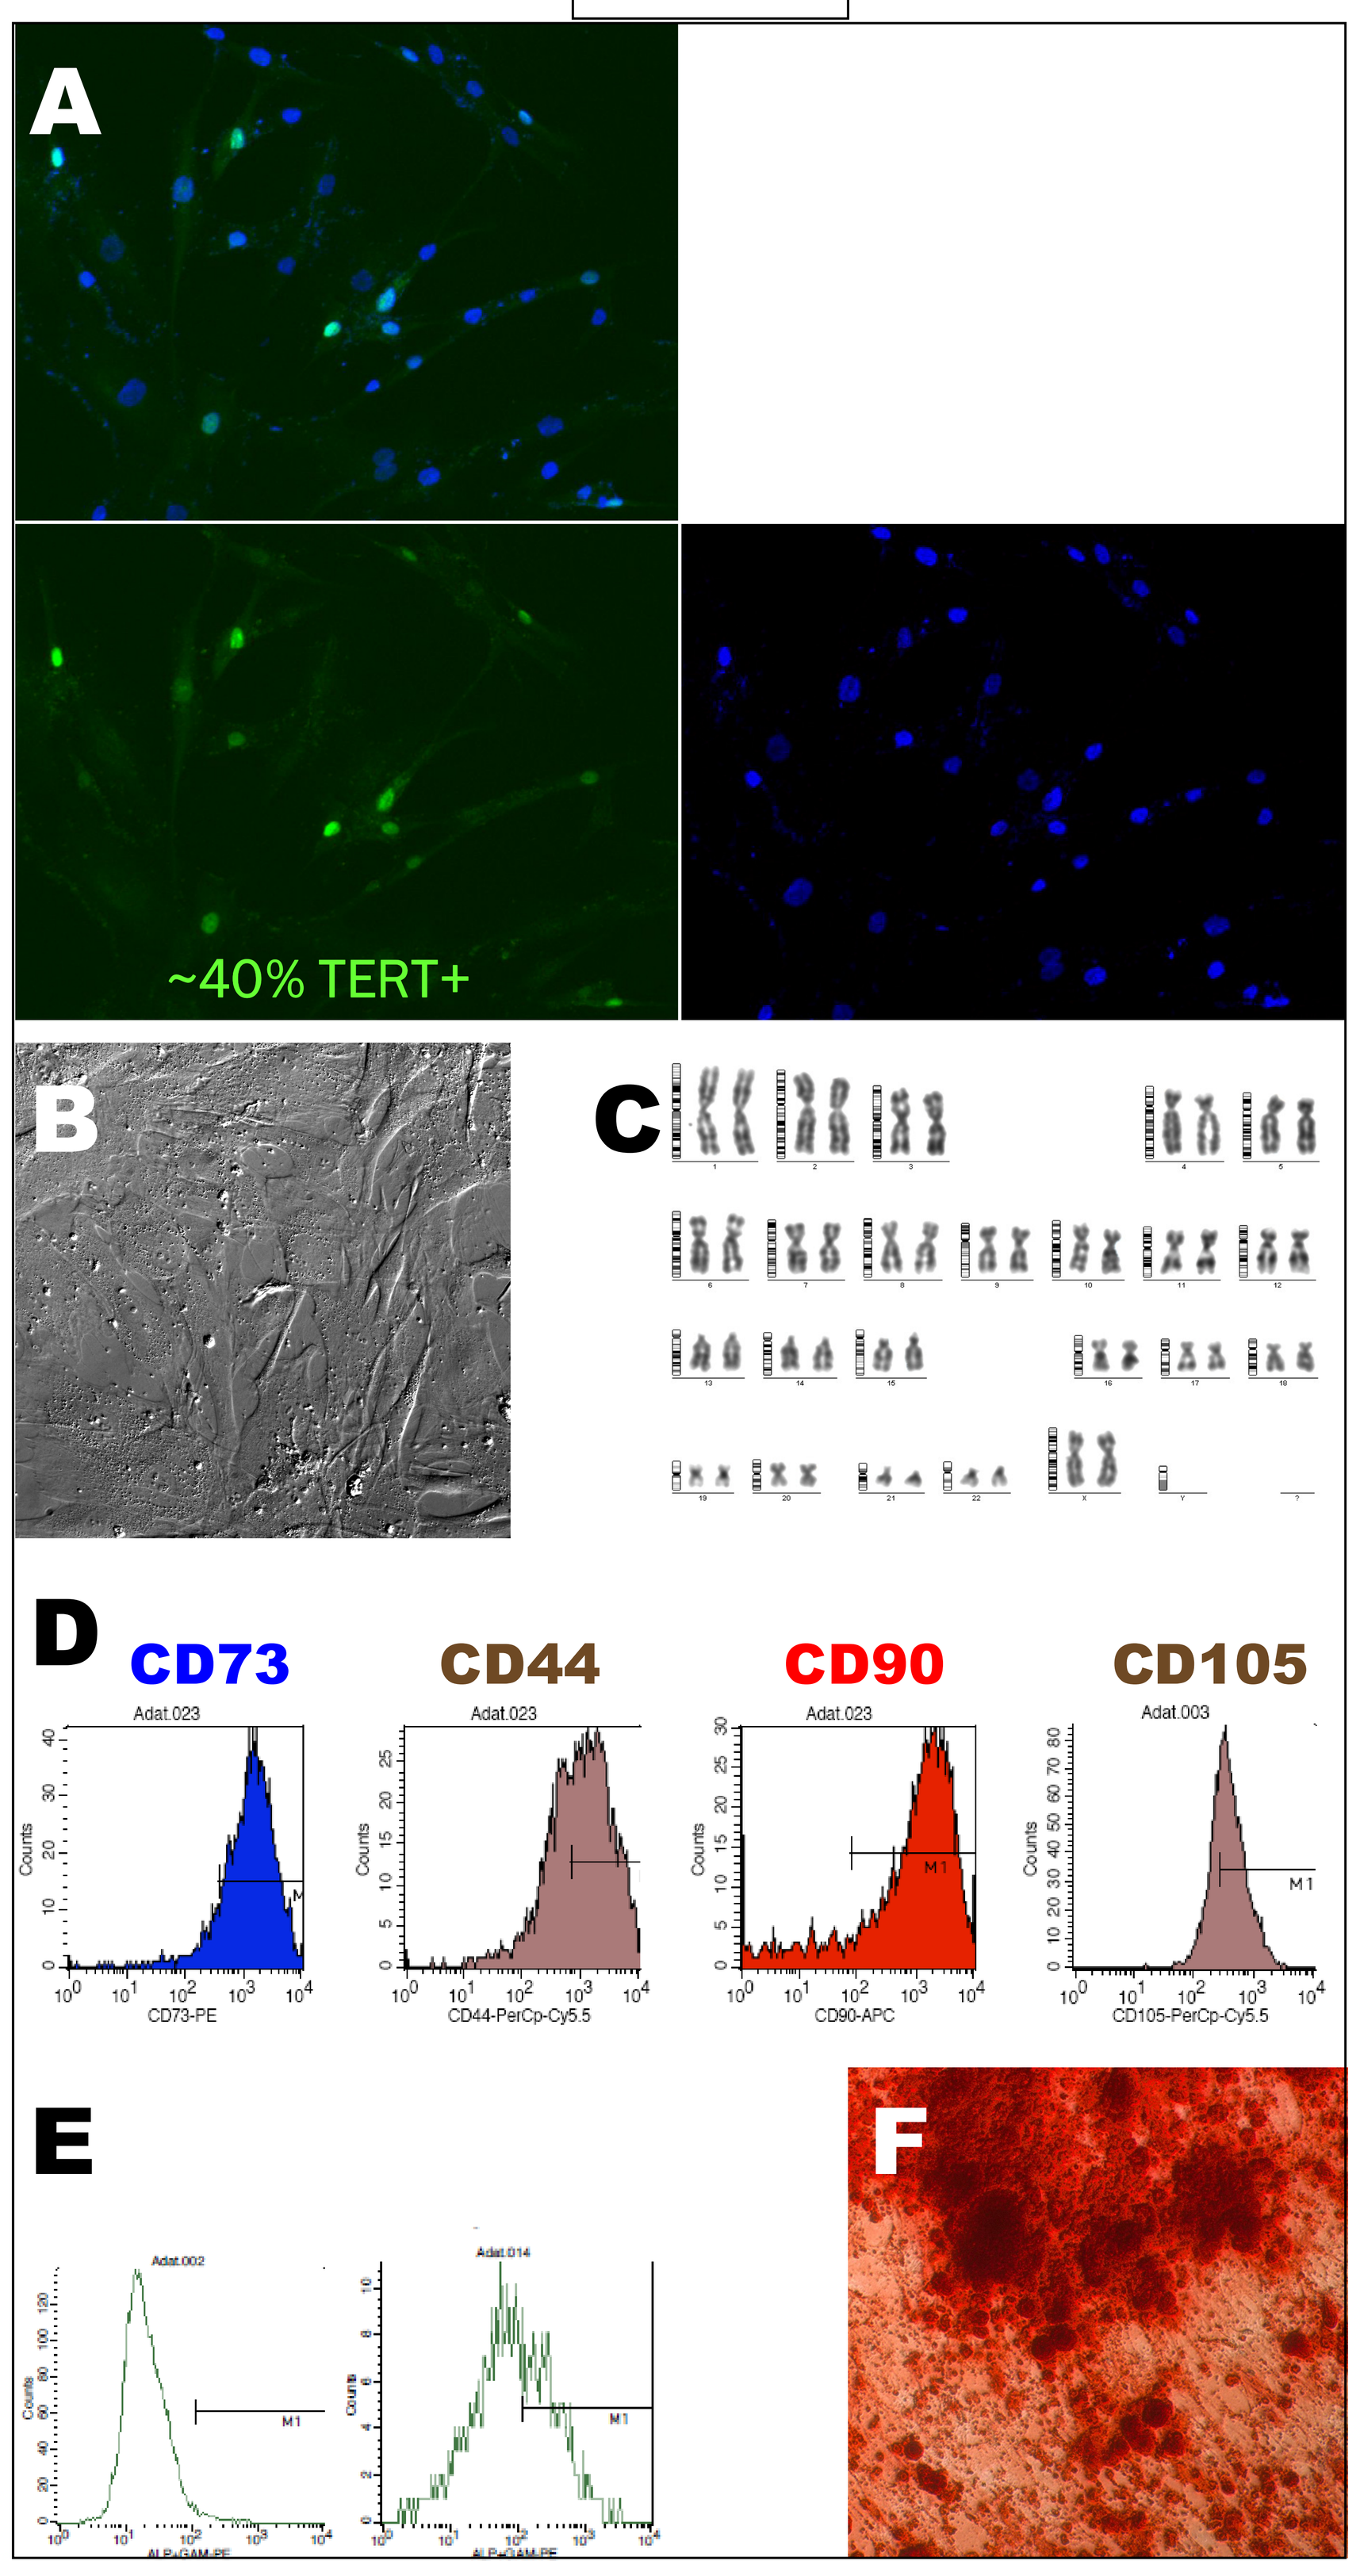
A
~40% TERT+
B
C
D
CD73
CD44
CD90
CD105
E
F

Bmi+TERT

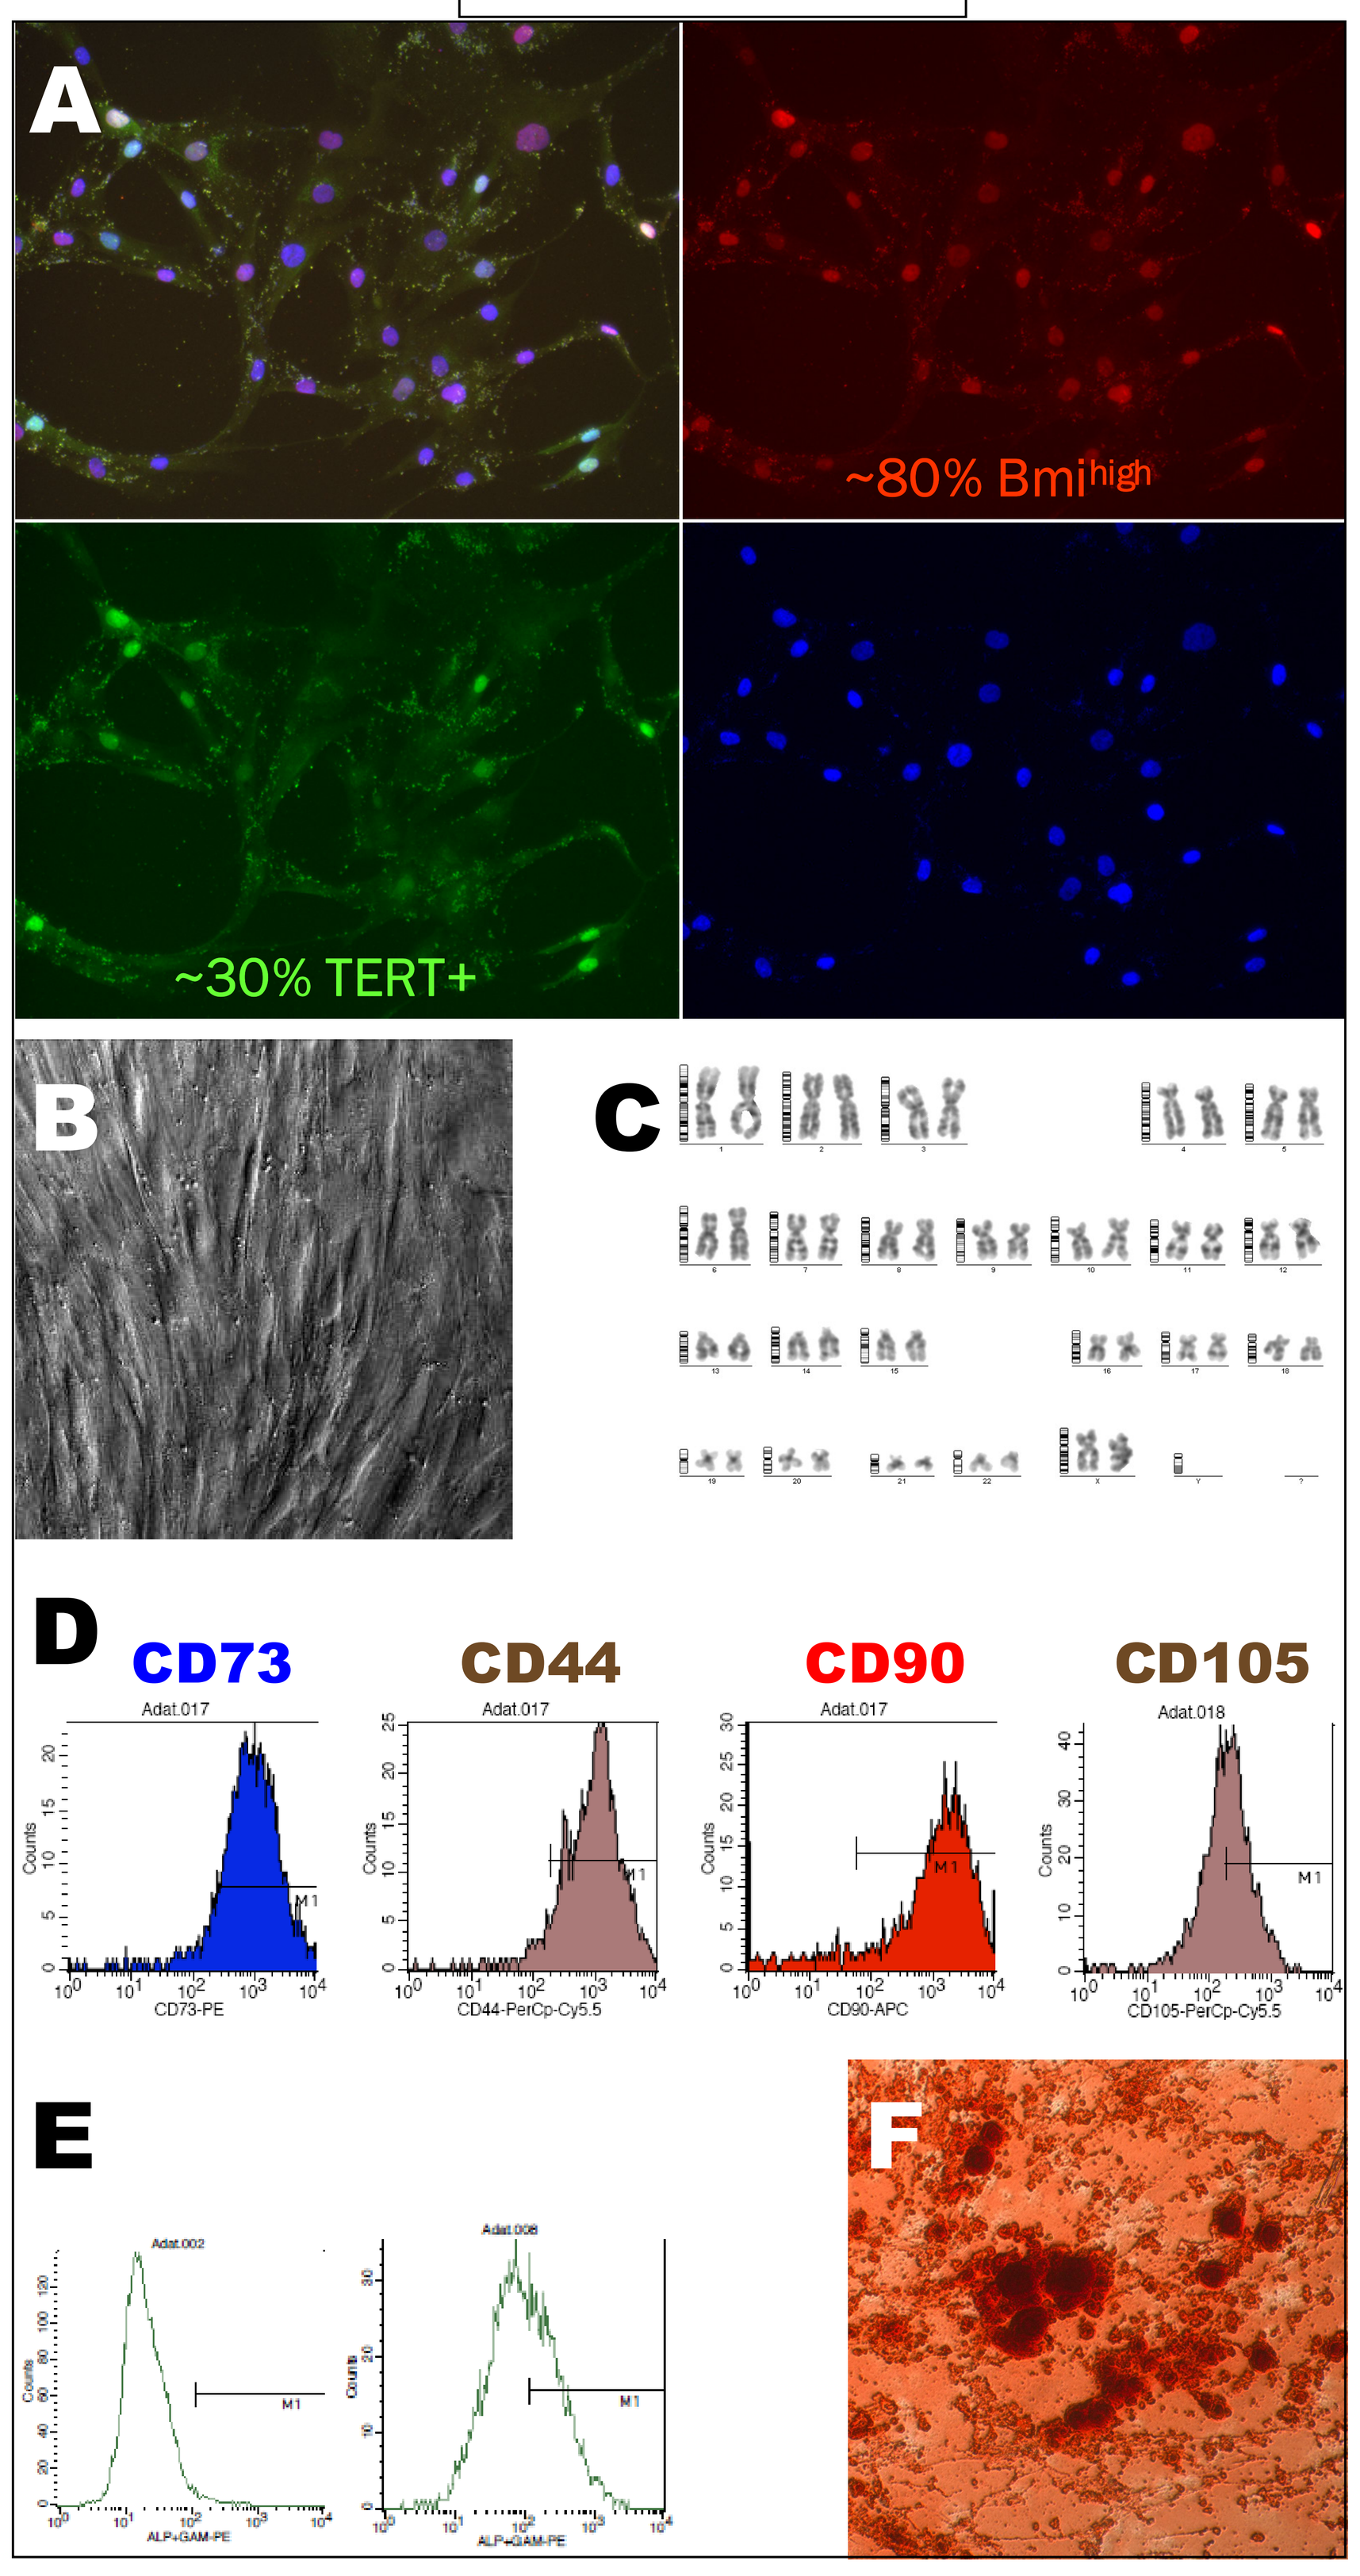
A
~80% Bmi^high
~30% TERT+
B
C
D
CD73
CD44
CD90
CD105
E
F

TAg+TERT

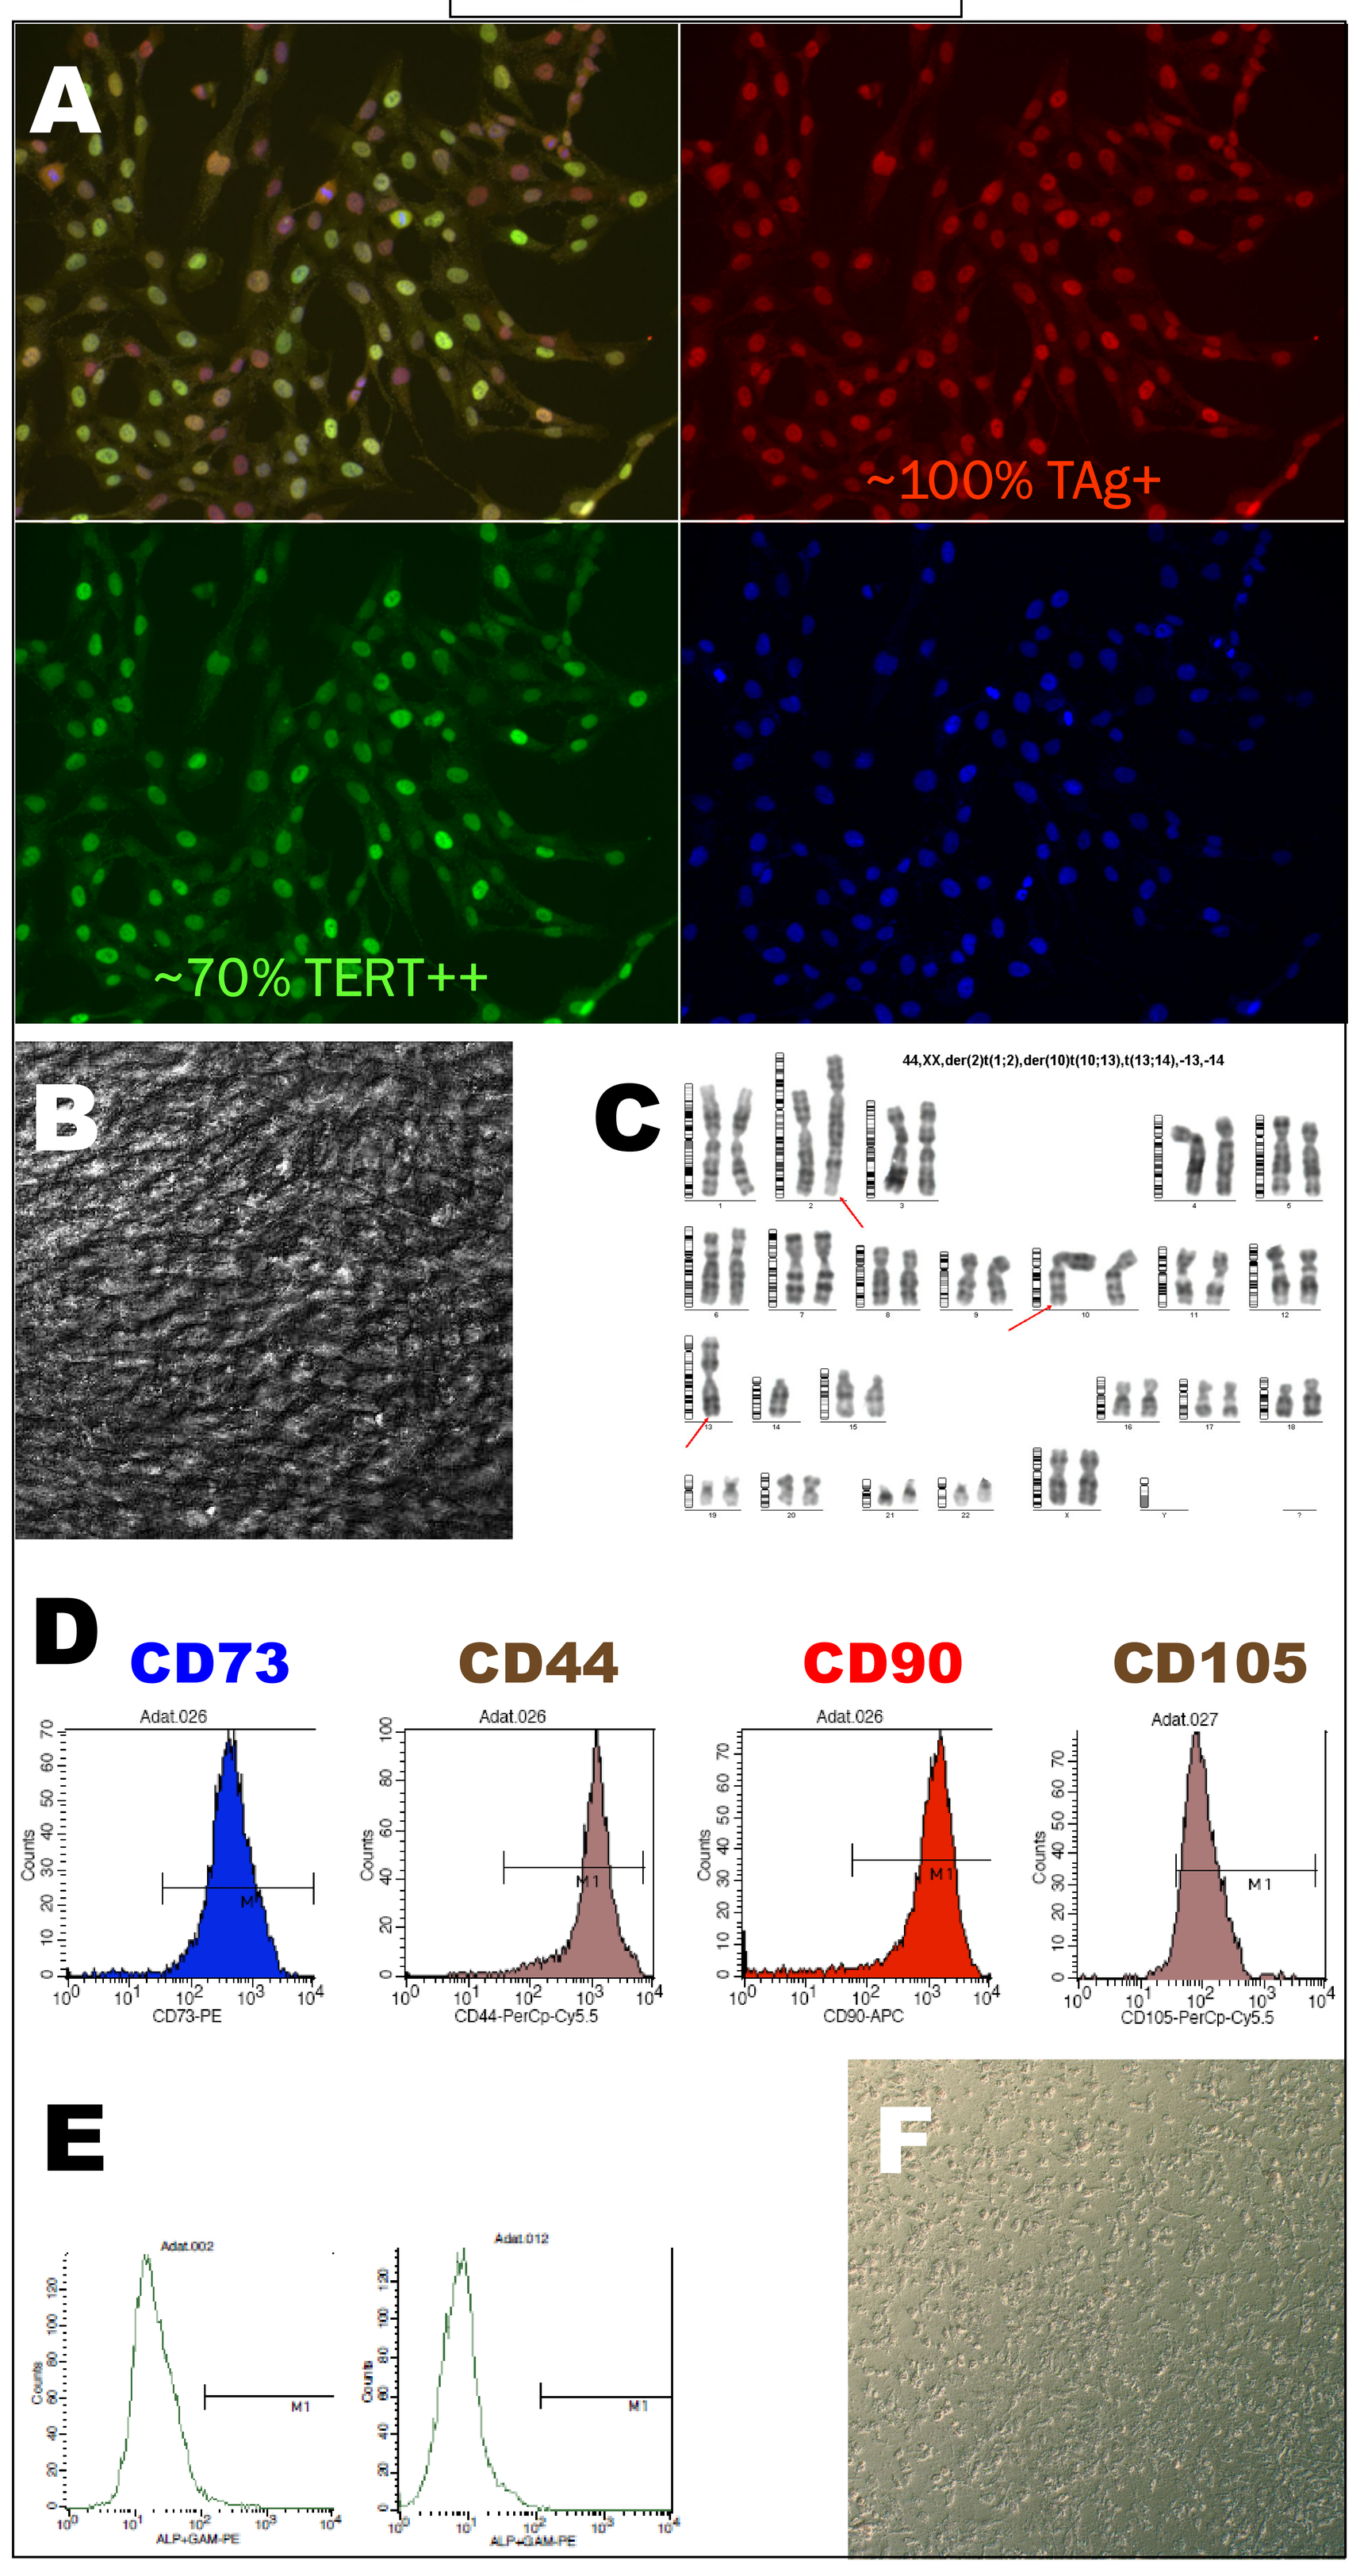
A
~100% TAg+
~70% TERT++
B
C
44,XX,der(2)t(1;2),der(10)t(10;13),t(13;14),-13,-14
D
CD73
CD44
CD90
CD105
E
F

Experimental Procedures and Results. The immortalizing gene human telomerase / reverse transcriptase (TERT), alone or in combination with the oncogenes Bmi-1 or SV40 large T antigen (TAg), was introduced by Cre/LoxP-excisable lentiviral vectors (Fig.1) into human adipose tissue-derived MSCs from a single donor. Cre-mediated excision of the transgene was tested in LoxP-GFP-expressing MSCs (Fig. 2).

The resulting transgenic populations TERT, Bmi+TERT and TAg+TERT, as well as the parental wild type MSC, were characterized by: A, immunofluorescence; red and green: transgenes as indicated, blue: DAPI; B, phase contrast microscopy; C, karyotyping; D, flow cytometry for standard MSC markers. Integrated proviral copy numbers were determined by qPCR (see Table 1).

Expression of the transgenes was also verified by RT-qPCR (Fig.3). Telomerase activity was measured by TRAPeze assay (Fig.4). Proliferation rate was assessed by resazurin conversion assay (Fig.5).

Finally, wild type and immortalized MSCs were subjected to osteogenic differentiation. E, Alkaline phosphatase (ALP) upregulation as measured by FACS, left: control WtMSC (no osteogenic diff.), right: osteogenic medium for 28 days. F, Alizarin red staining after 28 days of osteogenic induction.

Results are summarized in Table 1.

Fig.1

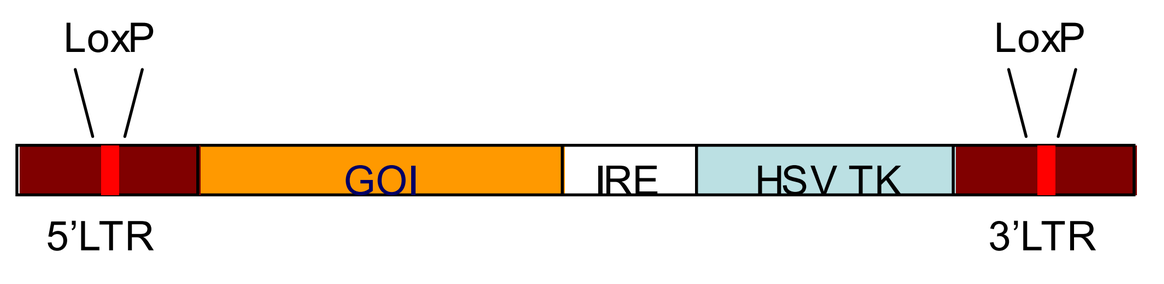
LoxP
LoxP
GOI
IRE
HSV TK
5'LTR
3'LTR

GOI, gene of interest
HSV TK: *Herpes simplex virus* thymidine kinase

Fig.2

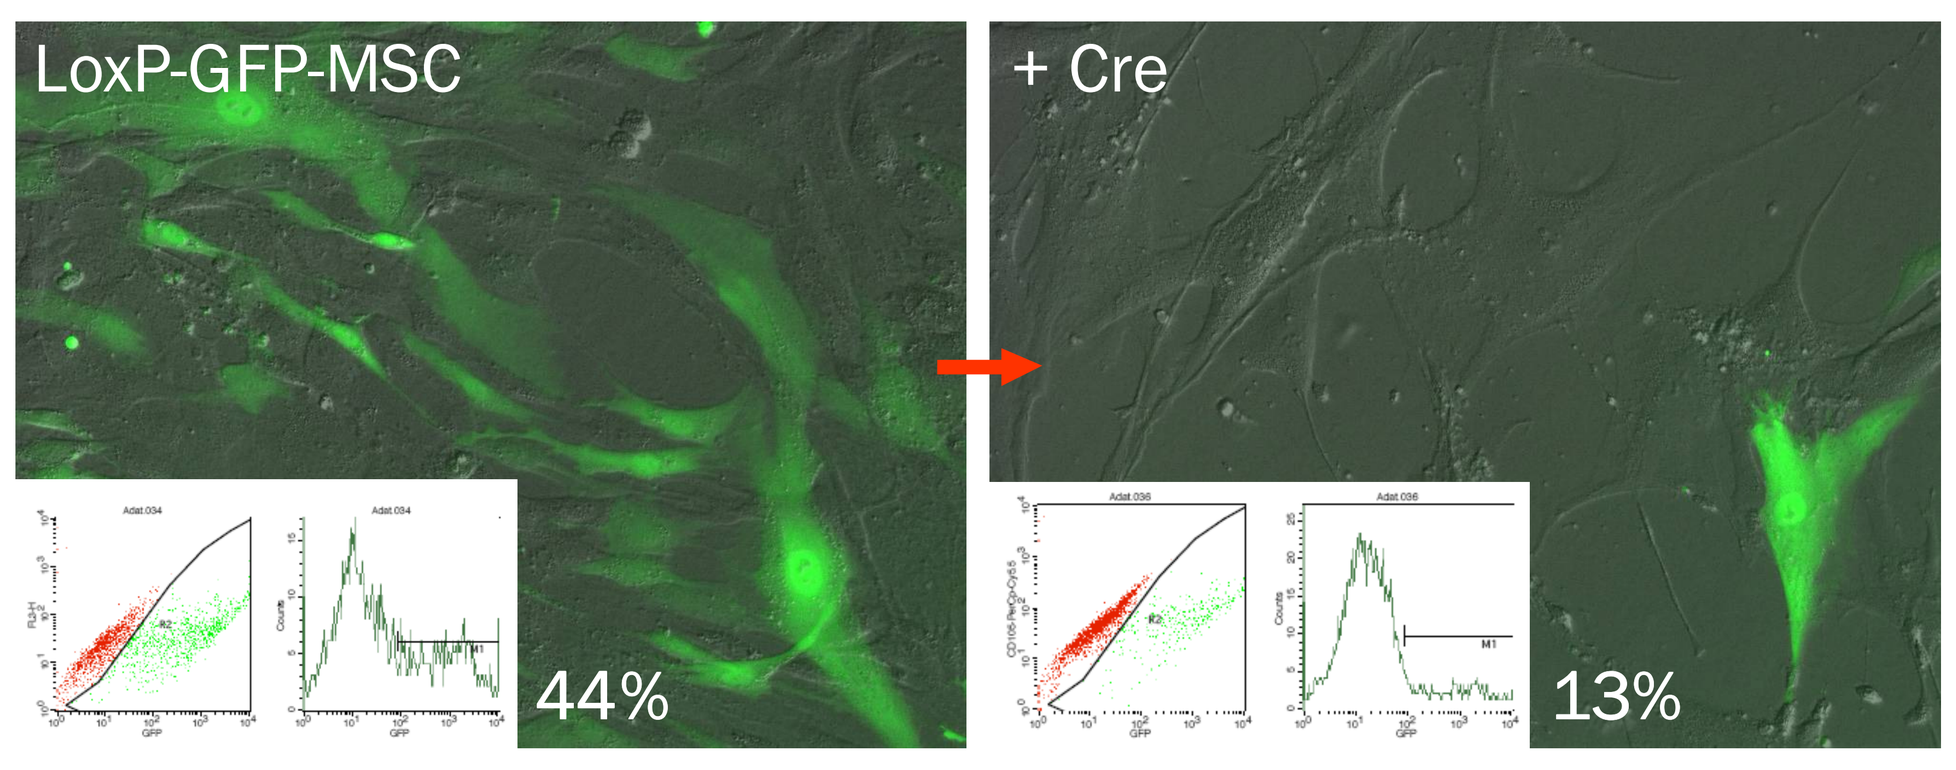
LoxP-GFP-MSC
+ Cre
44%
13%

Fig.3

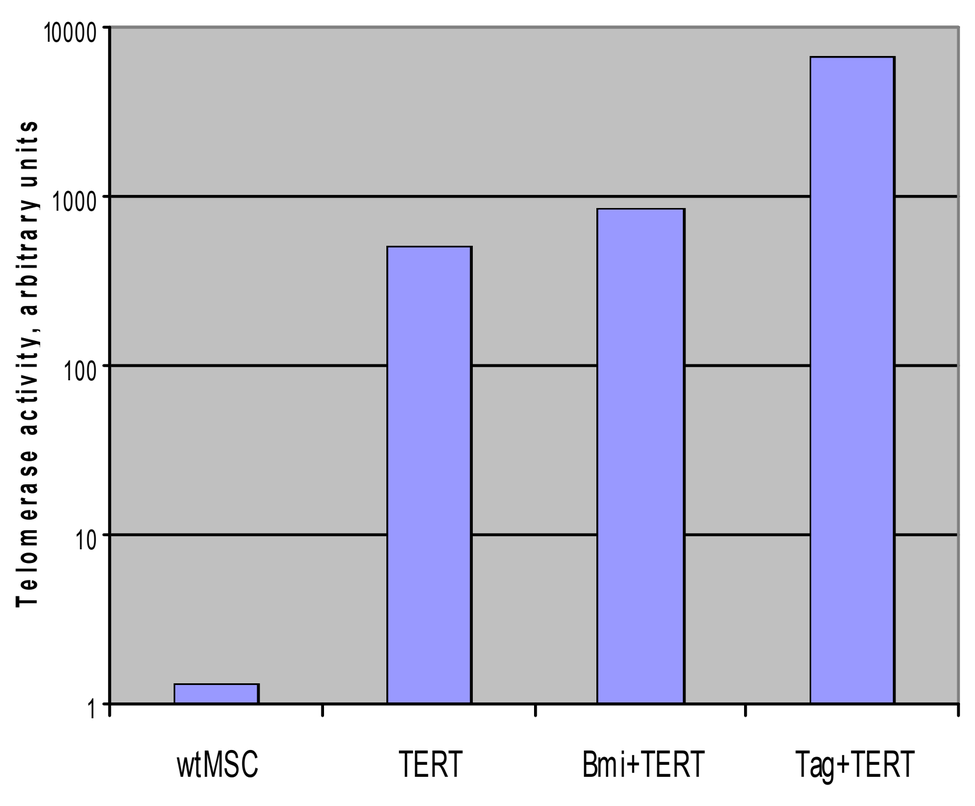
Telomerase activity, arbitrary units
wtMSC
TERT
Bmi+TERT
Tag+TERT

Telomerase activity

Fig.4

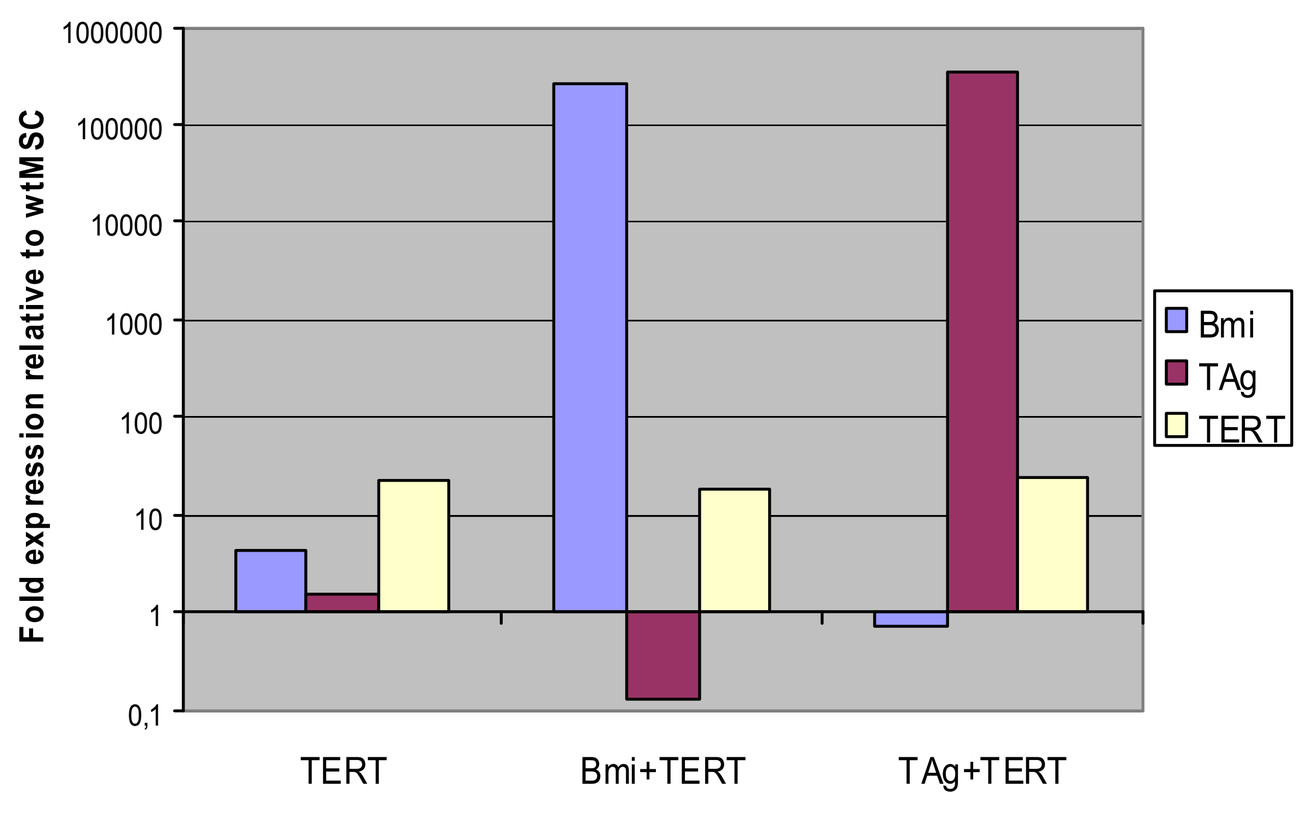
Fold expression relative to wtMSC
TERT
Bmi+TERT
TAg+TERT
Bmi
TAg
TERT

Transgene mRNA levels

Fig.5

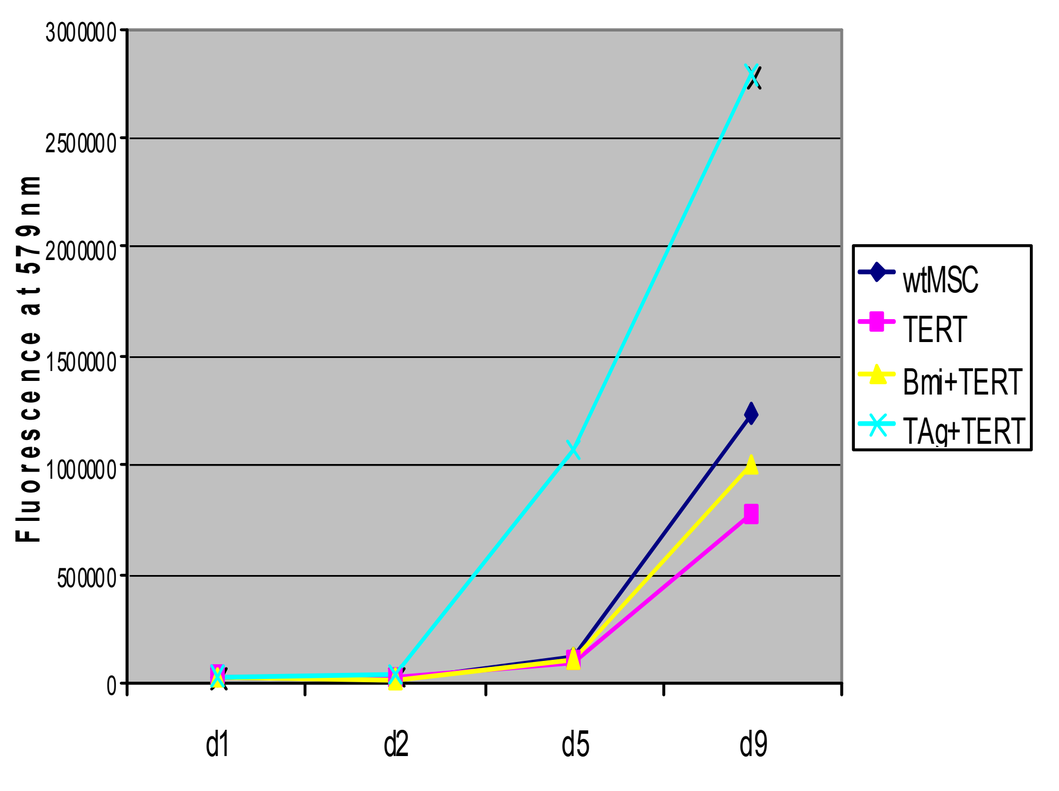
Fluorescence at 579nm
d1
d2
d5
d9
wtMSC
TERT
Bmi+TERT
TAg+TERT

Proliferation

Table 1

		WT MSC	TERT	Bmi+TERT	TAg+TERT
Phenotype	Microscopic phenotype	fibroblastic	like WT	like WT	small, rounded
	Proliferation	control	like WT	like WT	rapid
	MSC markers	all positive	all positive	all positive	all positive
	Hematopoietic markers	all negative (not shown)	all negative (not shown)	all negative (not shown)	all negative (not shown)
	Karyotype	normal	normal	normal	aberrant
Transgene	hTERT expression (IF)	undetectable	~40%	~30%	~70%
	Bmi-1 expression (IF)	low	low (not shown)	~80% Bmi^high	low (not shown)
	SV40 TAg expression (IF)	undetectable (not shown)	undetectable (not shown)	undetectable (not shown)	100%
	Telomerase activity	-	++	++	+++
	Average proviral copy number	0	not analyzed	6-7	3-4
Diff.	ALP induction	Y	Y	Y	N
	Calcification (Alizarin red staining)	Y	Y	Y	N

Conclusions. Since our cell lines have been subcultured over 30 times and show no sign of senescence, immortalization has probably been achieved. Bmi+TERT MSCs have maintained close-to-native MSC features and may be utilized in differentiation experiments. TAg+TERT cells, on the other hand, have become highly proliferative and lost differentiation potential. We are now exploring potential applications, as well as de-immortalization by removal of the transgenes.

Financial support: TÁMOP-4.2.1-IKUT from the National Development Agency (NFÜ).